©2024 Authors. Astellas is hereby authorized/granted a non-exclusive, perpetual license to use, reproduce, store, distribute, display, perform, and prepare derivative works based upon this publication or portions thereof to use for limited business purposes.

FPN: 2001P

Epidemiology and treatment patterns of patients with locally advanced or metastatic urothelial cancer in France: a non-interventional database study

Florence Joly[1–3], Morgan Rouprêt[4], Stéphane Culine[5,6], Aurore Tricotel[7], Emilie Casarotto[7], Rafaël Minacori[8], Torsten Strunz-McKendry[9], Khalil Karzazi[7], Kirsten Leyland[9], Marthe Vuillet[8], Marie-Catherine Thomas[8]

[1]INSERM, U1086 ANTICIPE, Normandie University, UNICAEN, Caen, France; [2]Clinical Research Department, Centre François Baclesse, Caen, France; [3]Medical Oncology Department, Centre François Baclesse de Caen, Caen, France; [4]Sorbonne University, GRC 5 Predictive Onco-Uro, AP-HP, Urology, Pitié-Salpêtrière Hospital, F-75013, Paris, France; [5]Department of Medical Oncology, Hôpital Saint-Louis, AP-HP, Paris, France; [6]Paris-Diderot University, France; [7]Real World Evidence Solutions, IQVIA, La Défense Cedex, France; [8]Astellas Pharma, Levallois-Perret, France; [9]Astellas Pharma Europe, Addlestone, United Kingdom

Objective

- To describe and assess the epidemiology and treatment patterns of patients with locally advanced or metastatic urothelial carcinoma (la/mUC) in France

Conclusions

- Incidences of la/mUC increased over time in France during the study period (from Jan 2020 to Dec 2022)
- Most patients who received first-line (1L) treatment from Jan 2020 to Jun 2022 had only chemotherapy as their 1L modality
 – More than 60% of patients only received 1L treatment
 – Avelumab use was low overall but increased over time (11.9–19.3%)
 – Only 17.7% of patients received second-line checkpoint inhibitors after 1L and 1.0% of patients received enfortumab vedotin
- The treatment landscape of la/mUC is further evolving with recent recommendations of enfortumab vedotin with pembrolizumab and of nivolumab with gemcitabine and cisplatin as 1L treatments[3]; further research is needed to assess the best treatment option in 1L according to the patient profile and the impact of subsequent treatments

References

1. Haute Autorite de Sante. Commission de la transparence. March 2021. https://www.hassante.fr/upload/docs/evamed/CT-18984_BAVENCIO_PIC_EI_AvisDef_CT18984.pdf (Accessed: 17 July 2024).
2. Haute Autorite de Sante. Évaluer les technologies de santé. December 2022. https://www.hassante.fr/upload/docs/evamed/CT19842_PADCEV_PIC_INS_AvisDef_CT19842.pdf (Accessed: 24 July 2024).
3. Powles T et al. *Ann Oncol*. 2024;35(6):485–490.

Conflicts of interest

Florence Joly received consulting fees from Astellas, Pfizer and Merck. **Morgan Rouprêt** received consulting fees from Ipsen, Astellas, Janssen, Astra Zeneca, Bayer and BMS. **Stéphane Culine** has no conflicts of interest to disclose. **Aurore Tricotel**, **Emilie Casarotto** and **Khalil Karzazi** are employees of IQVIA, contracted by Astellas Pharma Inc. to conduct the study. **Rafaël Minacori**, **Torsten Strunz-McKendry**, **Kirsten Leyland, Marthe Vuillet**, and **Marie-Catherine Thomas** are employees of Astellas Pharma Inc.

Acknowledgements

This study was sponsored by Astellas Pharma Inc. and Seagen, which was acquired by Pfizer Dec 2023. Medical writing support was provided by Pedro de Campos Silva, PhD, on behalf of Lumanity, funded by Astellas Pharma Inc. and Pfizer.

Electronic poster

Copies of this poster obtained through QR (Quick Response) and/or text key codes are for personal use only and may not be reproduced without written permission of the authors

For more information, please contact: Florence Joly, f.joly@baclesse.unicancer.fr

Presented at the European Society for Medical Oncology (ESMO) Annual Meeting, 13–17 September 2024, Barcelona, Spain and online

Background

- In France, the treatment landscape of la/mUC has recently changed
 – Avelumab was approved in 2021 as 1L maintenance treatment for patients with la/mUC who have not progressed after platinum-based chemotherapy[1]
 – Enfortumab vedotin was approved in 2022 for patients with la/mUC who have previously received platinum-based chemotherapy and a programmed death receptor-1 or programmed death-ligand 1 inhibitor[2]
- Here, we analyse the epidemiology and treatment patterns of la/mUC in France from 2020 to 2022

Methods

Study design

- EVOLVE-2 was a descriptive, non-interventional, longitudinal, and retrospective study
- Adult patients with la/mUC were identified from the Programme de Médicalisation des Systèmes d'Information, the French national database for hospitalisation records
- Data were extracted from 1 Jan 2015 to 31 Dec 2022

Study endpoints

- **Primary:**
 – Population 1: Annual incidence and prevalence
 – Population 2A: Annual incidence
- **Secondary:**
 – Population 3: patient characteristics and treatment patterns

Figure 1: Study flow chart – study population and sub-populations

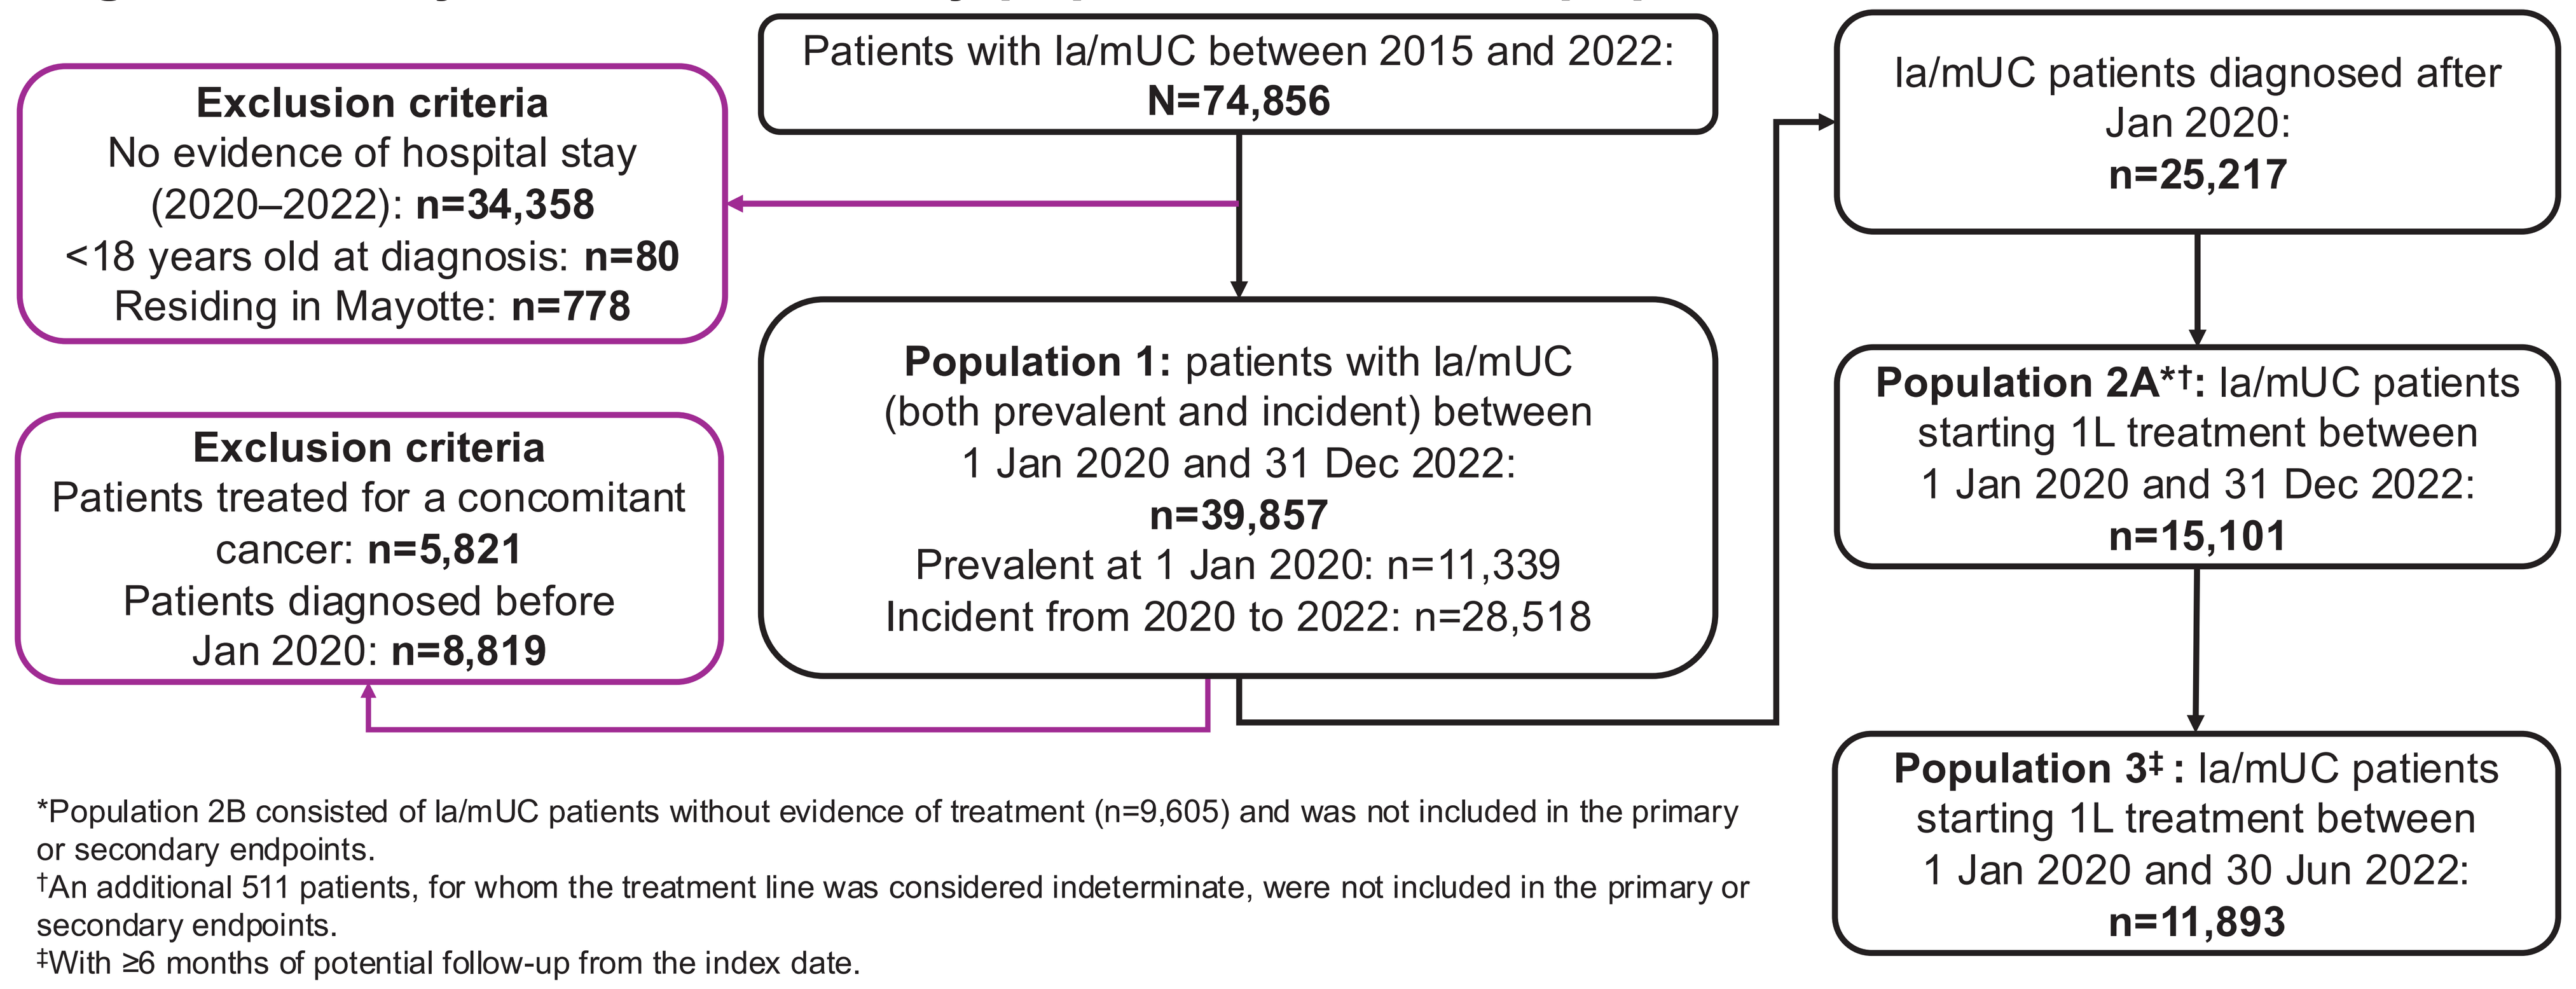
Patients with la/mUC between 2015 and 2022:
N=74,856
Exclusion criteria
No evidence of hospital stay (2020–2022): n=34,358
<18 years old at diagnosis: n=80
Residing in Mayotte: n=778
Population 1: patients with la/mUC (both prevalent and incident) between 1 Jan 2020 and 31 Dec 2022:
n=39,857
Prevalent at 1 Jan 2020: n=11,339
Incident from 2020 to 2022: n=28,518
Exclusion criteria
Patients treated for a concomitant cancer: n=5,821
Patients diagnosed before Jan 2020: n=8,819
la/mUC patients diagnosed after Jan 2020:
n=25,217
Population 2A*†: la/mUC patients starting 1L treatment between 1 Jan 2020 and 31 Dec 2022:
n=15,101
Population 3‡ : la/mUC patients starting 1L treatment between 1 Jan 2020 and 30 Jun 2022:
n=11,893

*Population 2B consisted of la/mUC patients without evidence of treatment (n=9,605) and was not included in the primary or secondary endpoints.
†An additional 511 patients, for whom the treatment line was considered indeterminate, were not included in the primary or secondary endpoints.
‡With ≥6 months of potential follow-up from the index date.

Results

- Of 39,857 patients with la/mUC, 25,217 (63.3%) were newly diagnosed from 1 Jan 2020 with no evidence of treatment for cancer other than UC after the date of la/mUC diagnosis (**Figure 1**)
- The prevalence and incidence of la/mUC ranged from 38.2 to 41.6 and 16.8 to 19.2 cases/100,000 people, respectively, from 2020 to 2022 (Population 1; **Figures 2A and 2B**)
- Treated incidence of la/mUC ranged from 7.9 to 11.2 cases/100,000 people from 2020 to 2022 (Population 2A, **Figure 2C**)
- Incidences were approximately four-fold higher in men than women and were higher in older (≥60 years) versus younger patients (**Figures 2B and 2C**)

Figure 2: Prevalence and incidence of la/mUC in France (2020–2022)

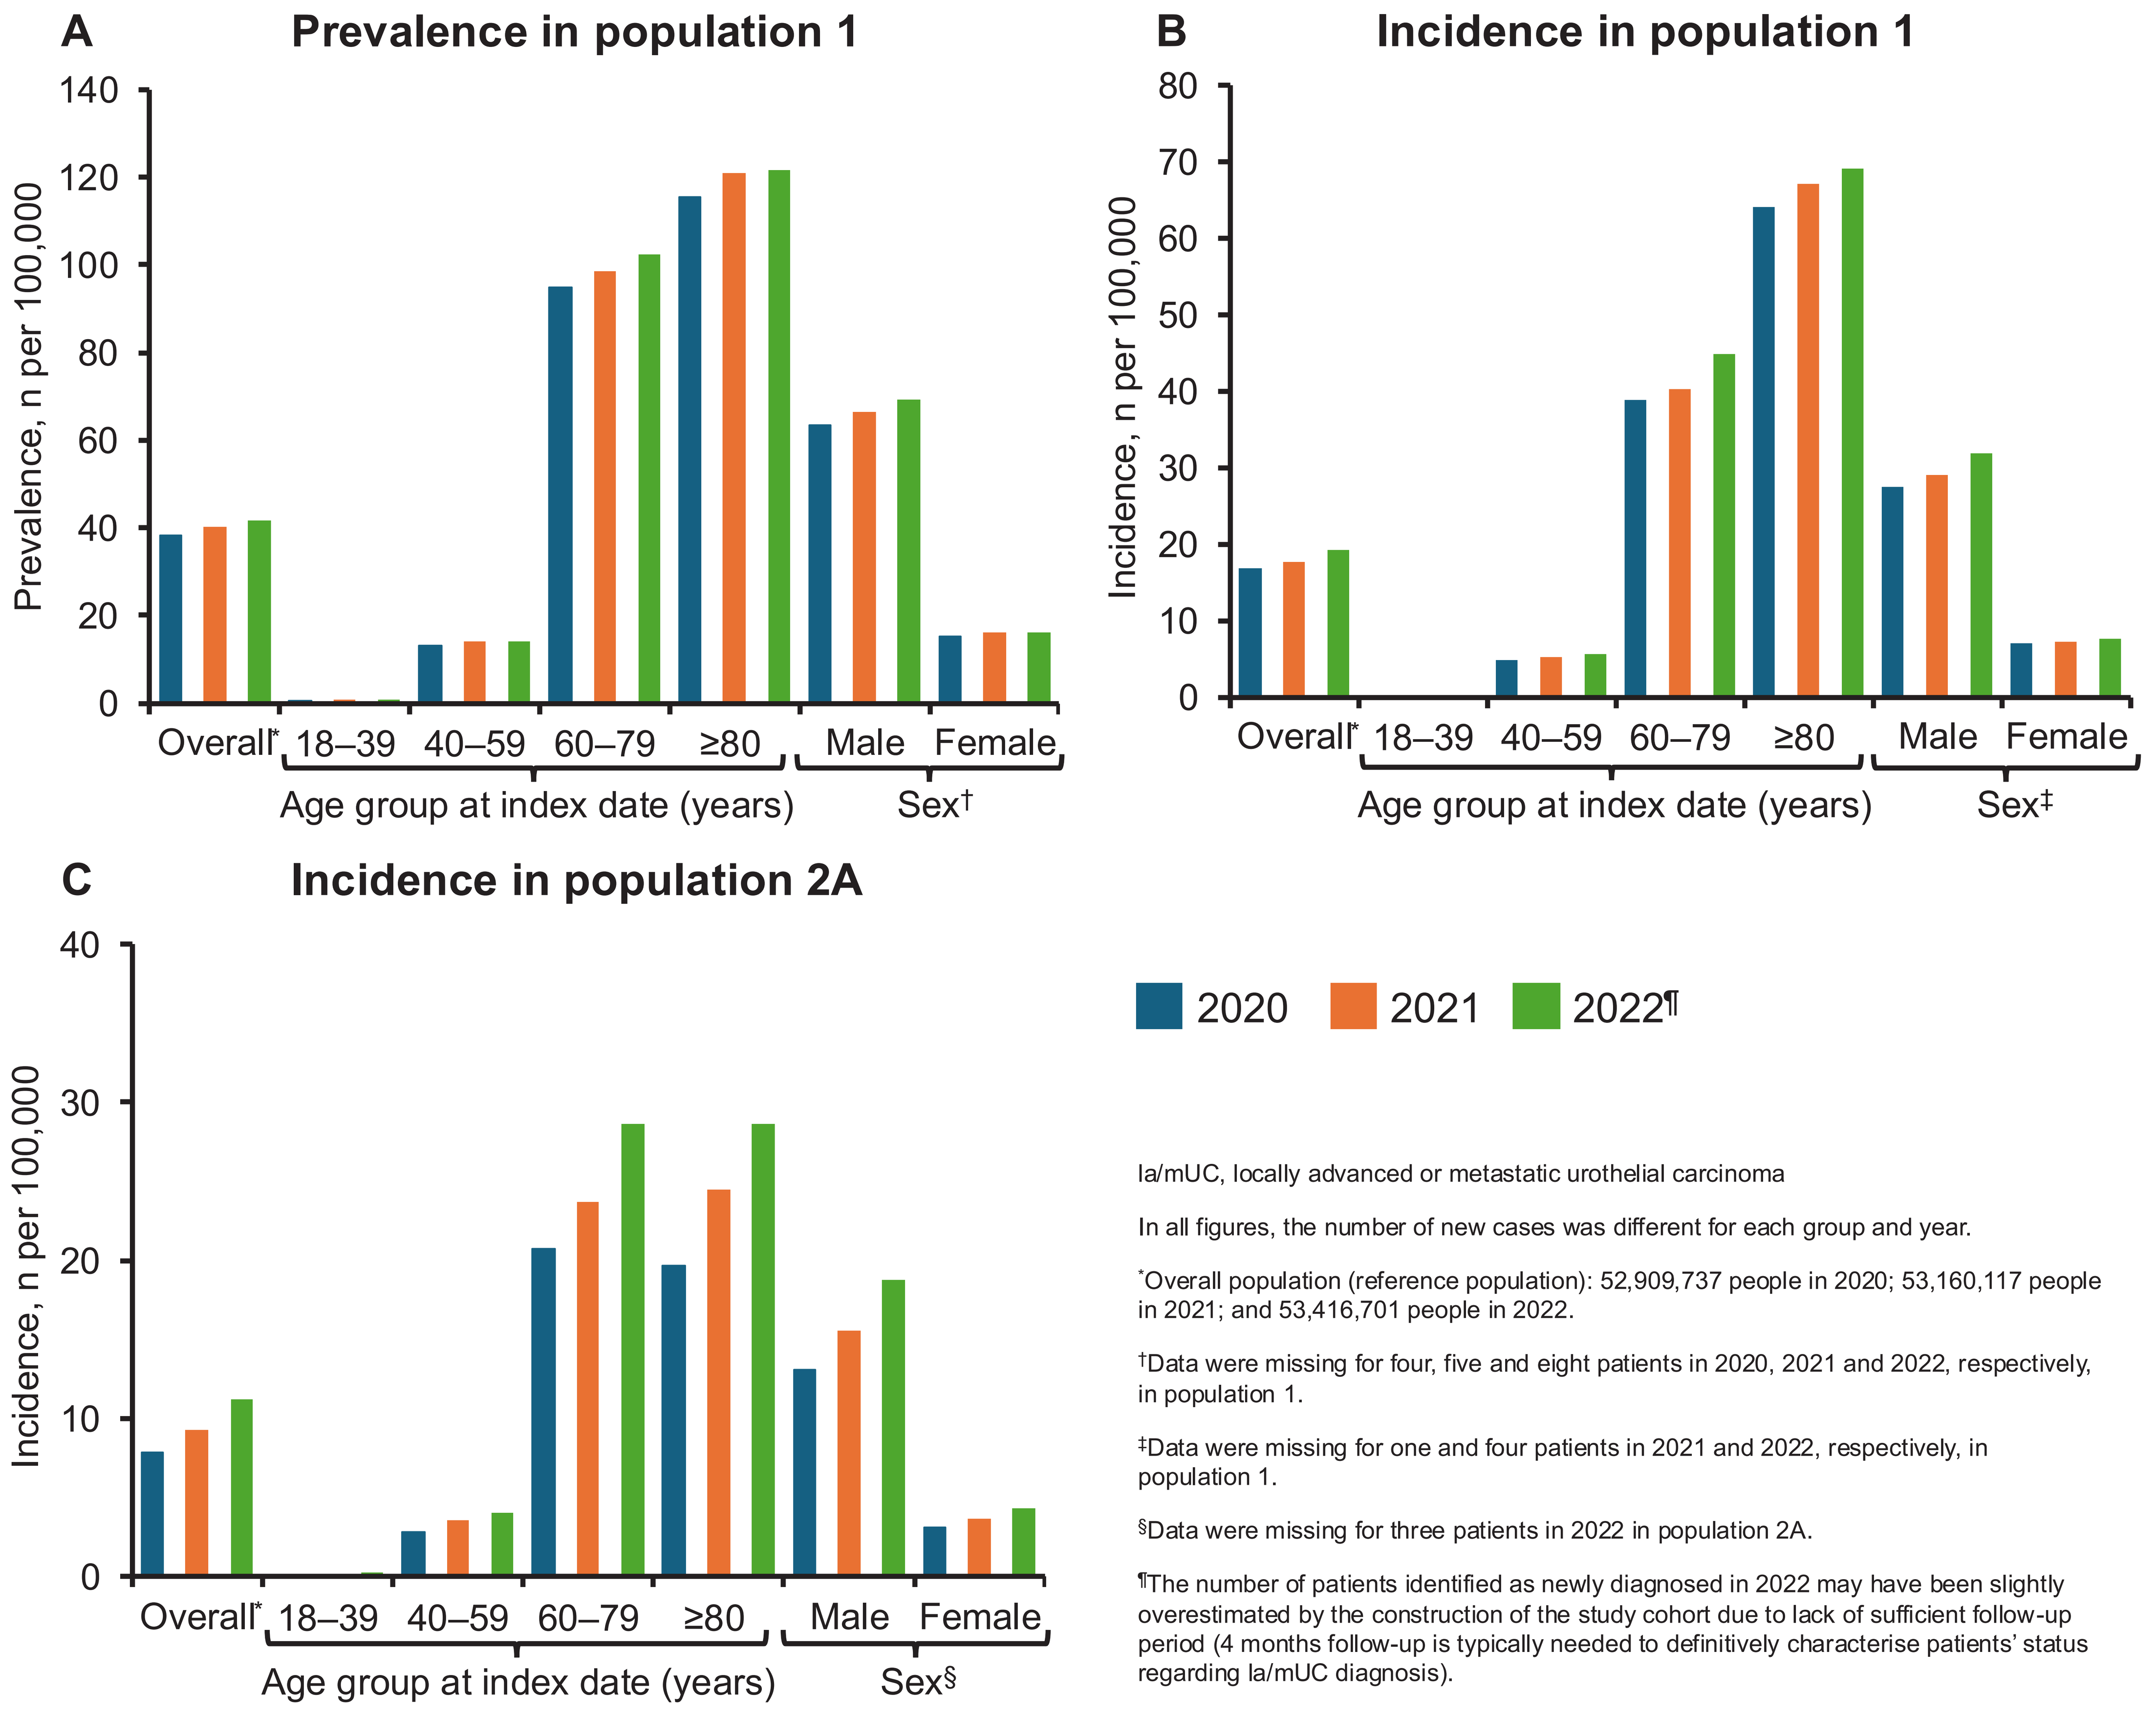
A
Prevalence in population 1
Prevalence, n per 100,000
140
120
100
80
60
40
20
0
Overall*
18–39
40–59
60–79
≥80
Male
Female
Age group at index date (years)
Sex†
B
Incidence in population 1
Incidence, n per 100,000
80
70
60
50
40
30
20
10
0
Overall*
18–39
40–59
60–79
≥80
Male
Female
Age group at index date (years)
Sex‡
C
Incidence in population 2A
Incidence, n per 100,000
40
30
20
10
0
Overall*
18–39
40–59
60–79
≥80
Male
Female
Age group at index date (years)
Sex§
2020
2021
2022¶

la/mUC, locally advanced or metastatic urothelial carcinoma

In all figures, the number of new cases was different for each group and year.

*Overall population (reference population): 52,909,737 people in 2020; 53,160,117 people in 2021; and 53,416,701 people in 2022.

†Data were missing for four, five and eight patients in 2020, 2021 and 2022, respectively, in population 1.

‡Data were missing for one and four patients in 2021 and 2022, respectively, in population 1.

§Data were missing for three patients in 2022 in population 2A.

¶The number of patients identified as newly diagnosed in 2022 may have been slightly overestimated by the construction of the study cohort due to lack of sufficient follow-up period (4 months follow-up is typically needed to definitively characterise patients' status regarding la/mUC diagnosis).

- In population 3, the mean (standard deviation) age of patients was 71.0 (9.8) years, and 79.5% of patients were male
 – The most common comorbidities (≥10%) at index date were other cancers (including lung and prostate cancers), peripheral vascular disease, chronic pulmonary disease, moderate or severe renal disease and myocardial infarction
- There were 64.5% of patients who received only 1L treatment in population 3
 – Almost all (93.6%) received platinum-based chemotherapy as 1L treatment (**Figure 3**)
- Overall, 17.0% of patients received subsequent avelumab maintenance therapy in population 3:
 – 11.9% (497/4,163) in 2020; 19.9% (986/4,947) in 2021; and 19.3% (536/2,783) in 2022

Figure 3: Sankey diagram of treatment patterns in population 3*†

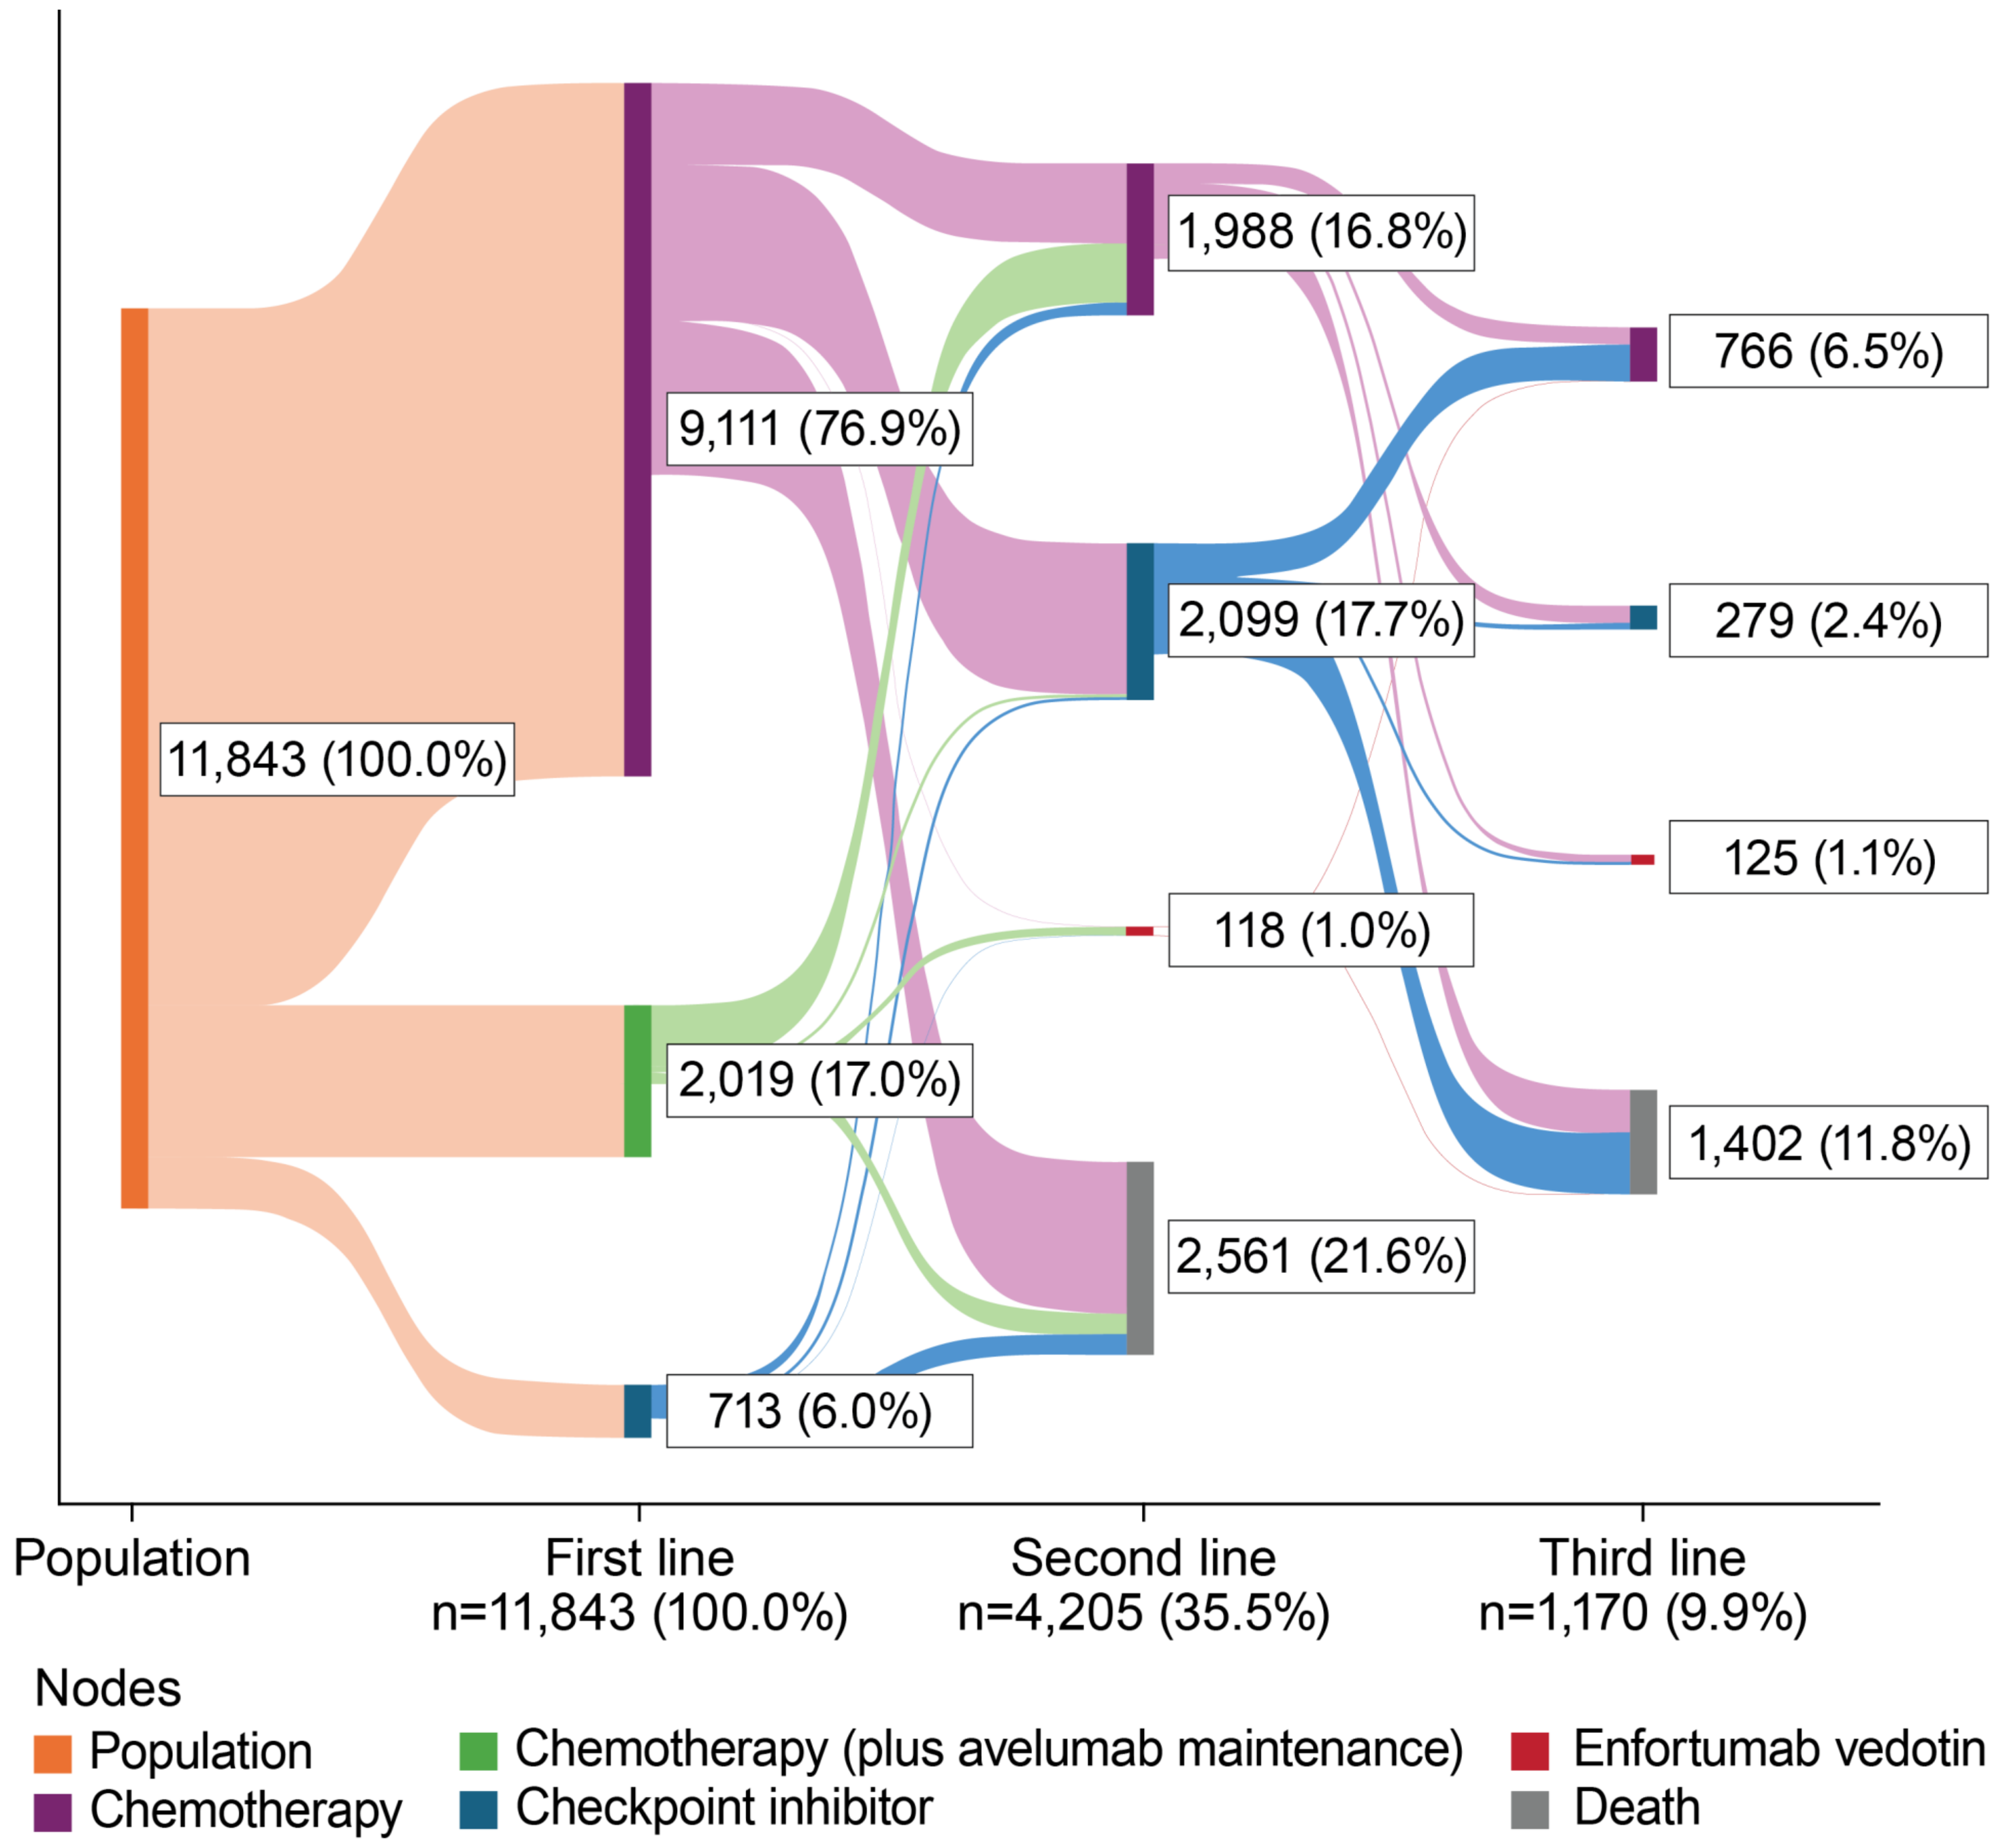
11,843 (100.0%)
9,111 (76.9%)
2,019 (17.0%)
713 (6.0%)
1,988 (16.8%)
2,099 (17.7%)
118 (1.0%)
2,561 (21.6%)
766 (6.5%)
279 (2.4%)
125 (1.1%)
1,402 (11.8%)
Population
First line
n=11,843 (100.0%)
Second line
n=4,205 (35.5%)
Third line
n=1,170 (9.9%)
Nodes
Population
Chemotherapy
Chemotherapy (plus avelumab maintenance)
Checkpoint inhibitor
Enfortumab vedotin
Death

*A total of 50 patients who received avelumab as 1L treatment without any evidence of prior chemotherapy were excluded from the analysis.
†The maximum follow-up period for incident patients was 3 years and patients included more recently may have not had sufficient time to experience a relapse during the study period and then begin a subsequent line of treatment.

©2024 Authors. Astellas is hereby authorized/granted a non-exclusive, perpetual license to use, reproduce, store, distribute, display, perform, and prepare derivative works based upon this publication or portions thereof to use for limited business purposes.