Teclistamab in Relapsed Refractory Multiple Myeloma patients on Dialysis: a French experience

F LACHENAL[1*], P. LEBRETON [2*], S. BOUILLIE[3], GM PICA[4], H. AFTISSE[5], L. PASCAL[6] , L. MONTES[7], M. MACRO[8], N. JOHNSON[9], S. HAREL[10], M FERNANDEZ[11], B. DE RENZIS[12], B. LIOURE[13], A. LAZARETH[14], M. JAVELOT[3], C LOUNI[15], A. HUART[16], A. PERROT [17]

1 CH Nord Dauphiné, Bourgoin-Jallieu, 2. CH J. Monod, Montivilliers, 3. Janssen, Issy-les-Moulineaux, 4. CH Chambery, 5. CH Saint-Quentin, 6. Hôpital Saint Vincent de Paul, Lille, 7. CHU Amiens, 8. CHU Caen, 9. Hôpital Cochin, Paris, 10. Hôpital Saint Louis, Paris, 11. Janssen, USA, 12. CHU Clermont-Ferrand, 13. ICANS, Strasbourg, 14. CHU Lyon, 15. IFM, Paris, 16. CHU Rangueil Toulouse, 17. CHU Oncopole, Toulouse.

INTRODUCTION

Up to 50% of patients with multiple myeloma (MM) have renal impairment at diagnosis; about 5% require dialysis. The outcome of these patients (pts) remains significantly worse and associated with early death.[1]

Teclistamab, a humanized CD3/BCMA bispecific antibody, is an important new treatment for pts with relapsed / refractory MM (RRMM) after triple-class exposure, delivering deep and durable responses.

However, data are lacking regarding its use in hemodialysis RRMM patients (HD-RRMM), a specific population commonly excluded from clinical trials.

AIM

Here, we report safety and efficacy data from RRMM pts treated with teclistamab in a dialysis setting.

METHOD

This retrospective study included RRMM pts with end-stage renal failure requiring dialysis treated with teclistamab in the French Acces Precoce program.

Teclistamab was given once weekly at dose of 1.5 mg/kg subcutaneously after step-up doses of 0.06 mg/kg and 0.3 mg/kg. Premedication was administered before teclistamab according to EMA approved prescribing information, and infection prophylaxis and immunoglobulin (IgG) replacement therapy was given according to local guidelines.

Pts did not object to participating in the study, which was submitted to the French Health Data Hub.

RESULTS

Population: Between November 2022 and October 2023, 15 HD-RRMM pts were treated with teclistamab in French hospitals typically administered post dialysis. The median age was 68 years [58-83]. The median time since diagnosis was 6 years [2-9]. The median follow-up was 5,2 months [0-10]. The median number of previous lines of therapy was 4 [3-6]. Most of the pts had myeloma-related end-stage renal failure. The isotype of myeloma was free light chains in 73% of cases. Among 10 evaluable pts, 7 were considered as high-risk based on cytogenetics (# 4, 5, 6, 8, 10, 11, 12).

Dose escalation and time to dialysis: The second step-up dose (dose 2) of teclistamab was given after a median of 3 days [2-7] and the first full treatment dose (dose 3) was given after a median of 7 days [4-14]. Teclistamab was mainly administrated just after dialysis for 9 pts, on an non-dialysis day for 3 pts. All pts had hemodialysis excepted one (#4, peritoneal dialysis). One pt (#8) improved renal function, allowing dialysis to be discontinued.

Cytokine release syndrome (CRS): Grade 1 or 2 CRS occured in 11 pts (exclusively during the step-up schedule) treated with tocilizumab in 4 pts, and addition of dexamethasone in one pt (#3). No grade 3-4 was reported.

Immune effector cell-associated syndrome (ICANS): Grade 2 ICANS co-occured with grade 2 CRS in one pt (#15, 80 years old), both resolved after two doses of dexamethasone.

Objective response rate: Median time to best response was 40 days [6-124]. Thirteen pts achieved a response (2 sCR, 2 CR, 7 VGPR and 2 PR). The one progressive pt (#14) presented with extramedullary disease and died prematurely from sepsis. Patient #15 was not evaluable for response. (Figure 1)

Infection recurrence and prevention: Nine patients had grade ≥ 2 infectious complication. Seven pts experienced bacteriema (2 fatal cases; #14, #7); two pts presented a viral infection (#1, #3) (Figure 1). Infectious prophylaxis included sulfametoxazole - trimethoprim and valaciclovir (for 12 pts), as well as oracillin (for 6 pts). A primary prophylaxis with subcutaneous or intravenous polyvalent immunoglobulins (IgG) was given to 8 pts.

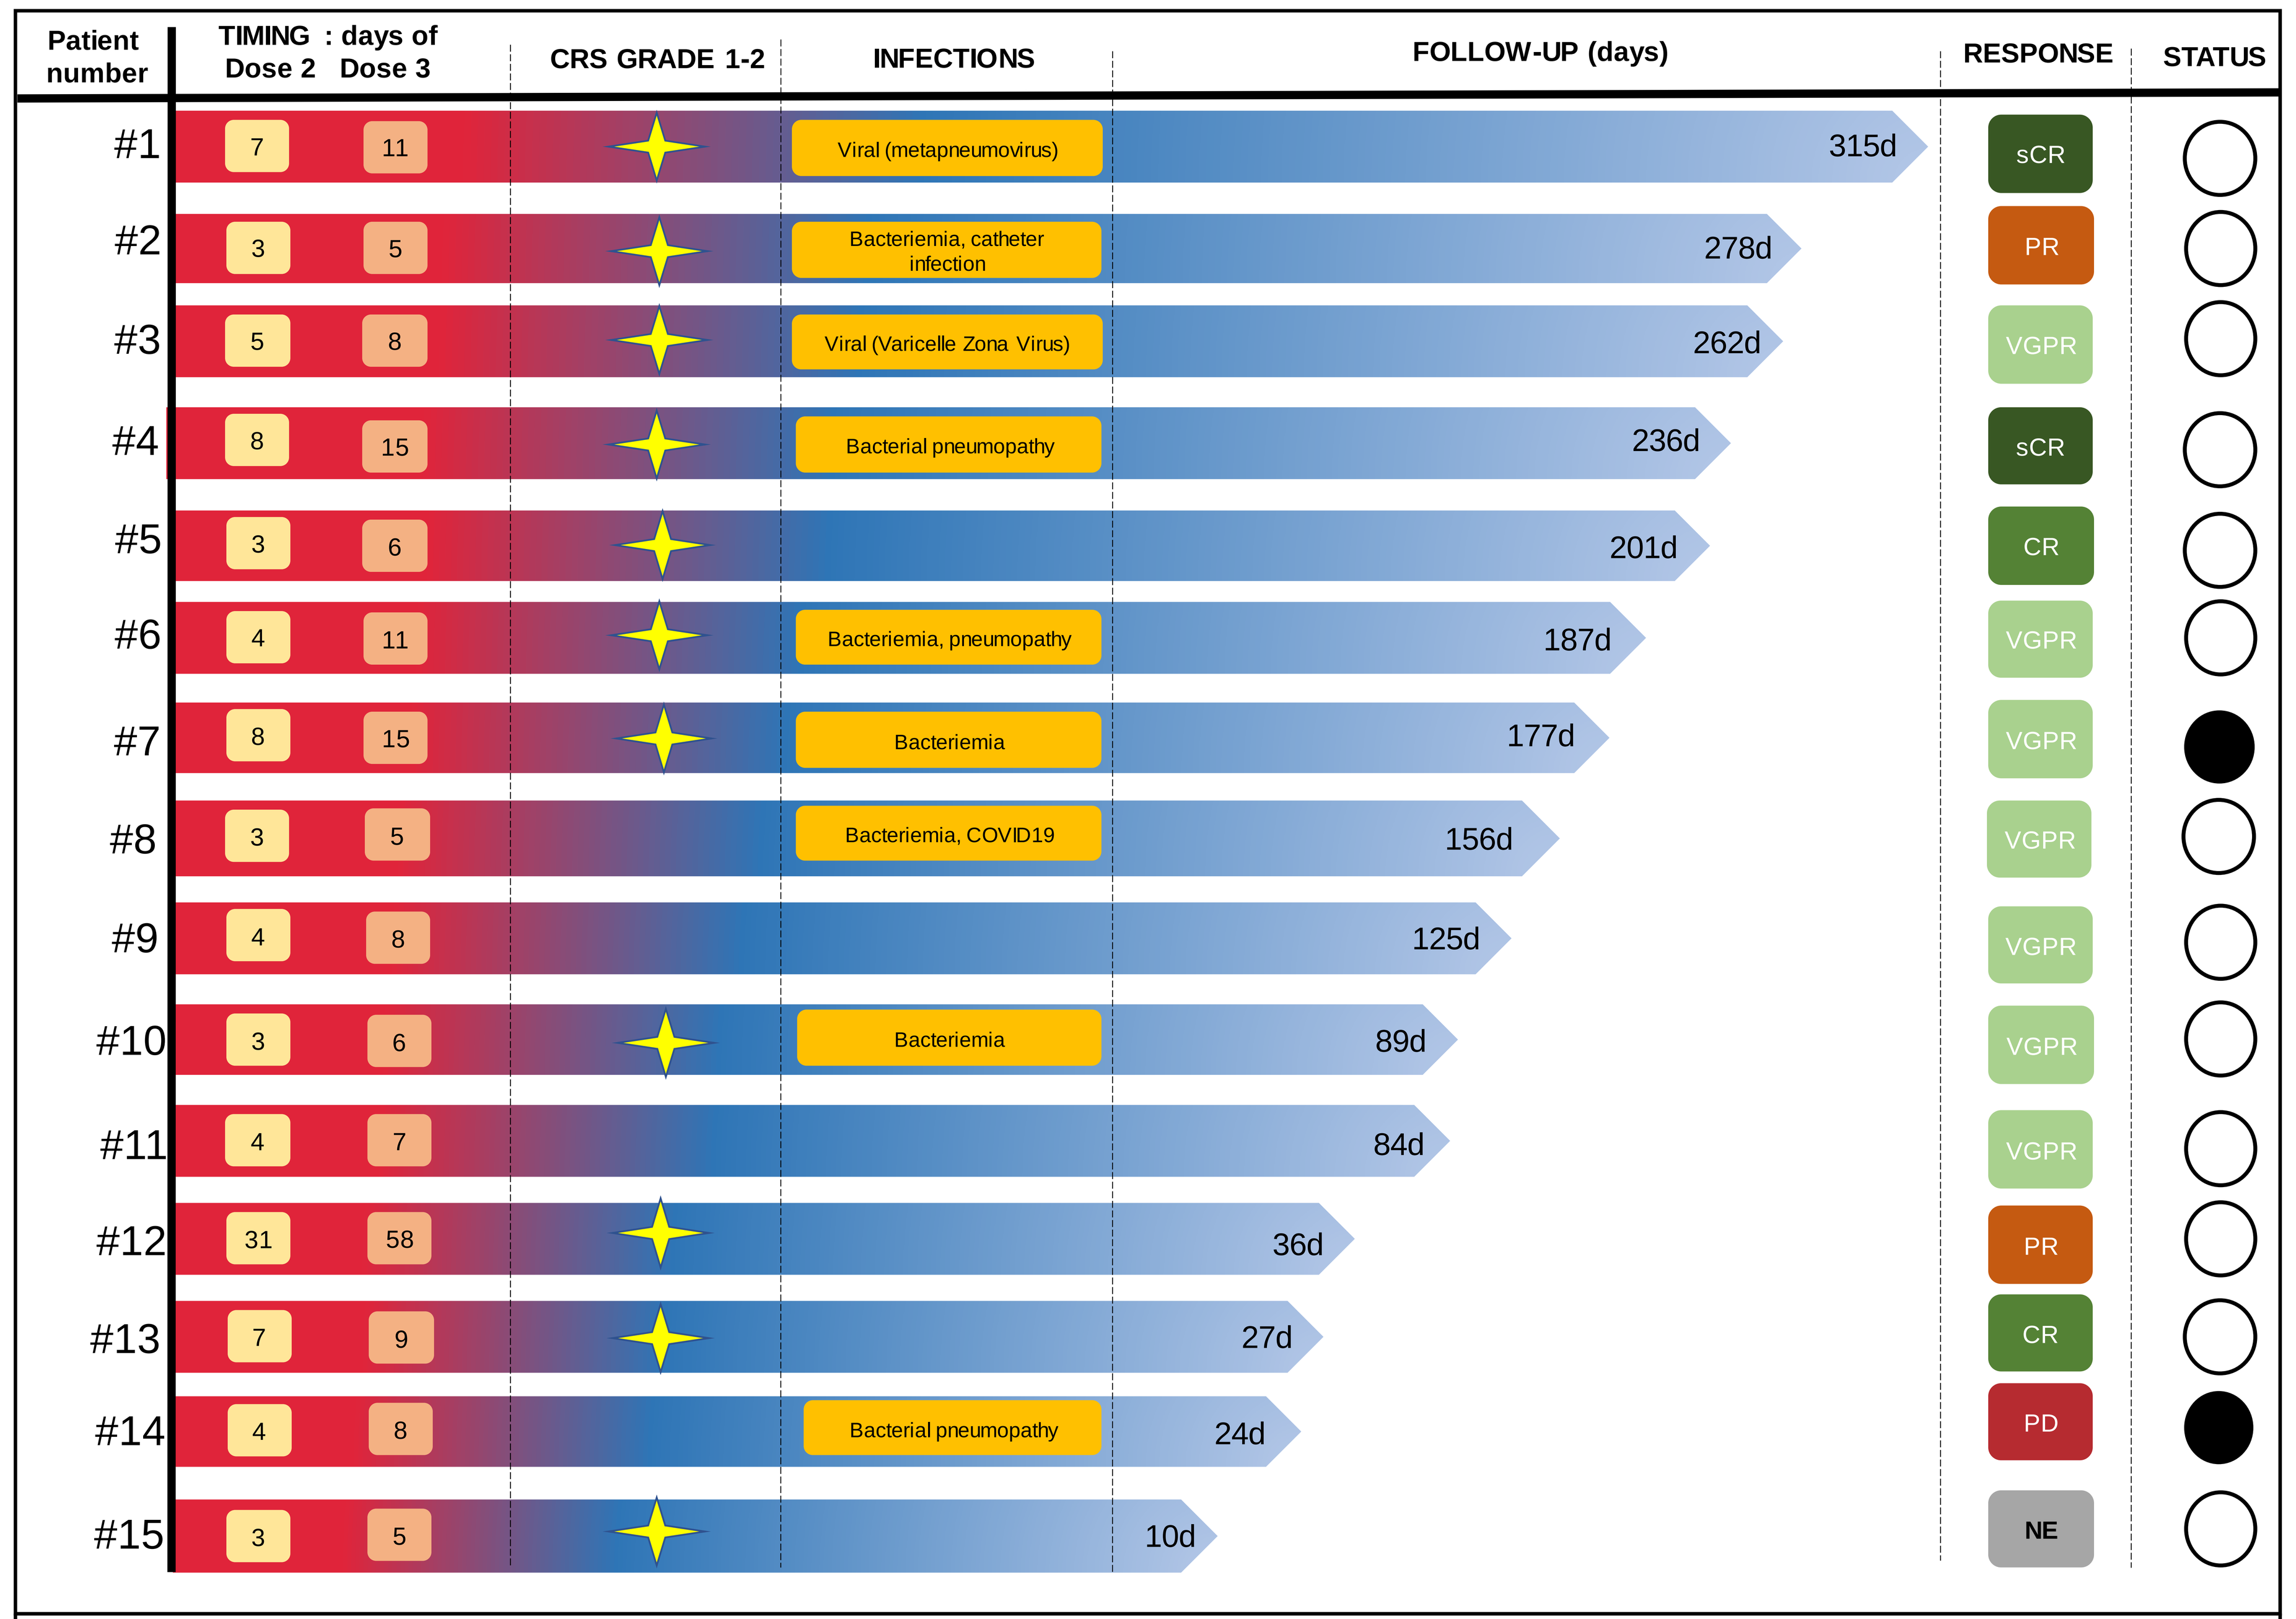
Patient number
TIMING : days of Dose 2 Dose 3
CRS GRADE 1-2
INFECTIONS
FOLLOW-UP (days)
RESPONSE
STATUS
#1 7 11 Viral (metapneumovirus) 315d sCR
#2 3 5 Bacteriemia, catheter infection 278d PR
#3 5 8 Viral (Varicelle Zona Virus) 262d VGPR
#4 8 15 Bacterial pneumopathy 236d sCR
#5 3 6 201d CR
#6 4 11 Bacteriemia, pneumopathy 187d VGPR
#7 8 15 Bacteriemia 177d VGPR
#8 3 5 Bacteriemia, COVID19 156d VGPR
#9 4 8 125d VGPR
#10 3 6 Bacteriemia 89d VGPR
#11 4 7 84d VGPR
#12 31 58 36d PR
#13 7 9 27d CR
#14 4 8 Bacterial pneumopathy 24d PD
#15 3 5 10d NE

Figure 1. Course of teclistamab treatment in HD-RRMM patients: step-up dose days (step-up dose 2 0.3 mg/kg; first full dose 1.5 mg/kg), CRS occurrence, infectious complication, last follow-up, International Myeloma Working Group response status, vital status (alive - dead).
NE: non evaluable; sCR: stringent complete response; CR: complete response; VGPR: very good partial response; PR: partial response; PD: progressive disease.

CONCLUSIONS

Despite limited experience from a small cohort, teclistamab monotherapy seems to be a feasible, effective and safe option for pts with HD-RRMM. The incidence of CRS and ICANs are comparable with those seen in MajesTEC-1.[2] Infections are the main complication, as reported by *Joiner et al*[3], which supports the systematic use of bacterial and viral prophylaxis, including IgG replacement therapy. These data are very important for these patients typically excluded from clinical trials and CAR-T cell therapies that usually require fludarabine-based lymphodepletion regimen (the dose of which is difficult to adjust with dialysis).

REFERENCES

1. Dimopoulos MA, Merlini G, Bridoux F et al. Management of multiple myeloma-related renal impairment: recommendations from the International. Myeloma Working Group. Lancet Oncol 2023; 24: e293–311
2. Moreau P, Garfall AL, van de Donk N, et al. Teclistamab in relapsed or refractory multiple myeloma. N Engl J Med. 2022;387(6):495-505.
3. Laura Joiner, et al. Teclistamab in patients with multiple myeloma and impaired renal function. Am J Hematol. 2023;98:E322–E324.

ACKNOWLEDGEMENTS

The authors wish to thank all the patients and their families, the centers that participated to the study and the IFM group.

CONTACT INFORMATION

perrot.aurore@iuct-oncopole.fr
pierre.lebreton@ch-havre.fr